Large-scale synchrony in alpha and gamma bands underlies attention to multiple visual objects

Santeri Rouhinen[1], J Matias Palva[1], Satu Palva[1].

1) University of Helsinki, Finland

Introduction

- The capacity of attention has been studied with multi object tracking (MOT) tasks in which subjects track the presence of one to n objects. These studies have revealed that individual capacity of attention varies between 3–4 objects (Vogel 2004, cf. Oksama 2004).
- fMRI studies with MOT tasks have shown that BOLD signal fluctuations in lateral prefrontal and posterior parietal cortices are positively correlated with the number of attended objects (Culham 1998, Jovicich 2001).
- Little is known of the mechanisms underlying the capacity of attention in fast temporal scales.
- Oscillations and synchronization integrate neuronal processing and regulate neural communication (Singer 1999, Fries 2005) and might hence also be a mechanism to limit the capacity of attention by regulating and controlling communication between brain areas. We have shown by using magneto- and electroencephalography (M/EEG) recordings and MOT task that the strength of local gamma oscillations is correlated positively with the capacity of attention (Rouhinen 2013).
- We now investigated whether the large-scale synchronization could be a neuronal mechanism to integrate anatomically distributed processing and to limit the capacity of visual multi-object attention.

Methods

- We used concurrent magneto- and electroencephalography (M/EEG) to record neuronal activity during a MOT task and data-driven analysis of inter-areal phase synchronization.
- M/EEG was recorded from nineteen healthy subjects who performed two visual multi-object tracking tasks with semi-naturally moving objects. In the task, subject tracked 1–4 target objects and detected brief (< 100 ms) feature changes. In Task1, the number of tracked objects varied from 1 to 4, while in the Task2, target objects were presented among 1–3 distracters.
- MaxFilter software and independent component analysis (ICA) were used to suppress extra-cranial noise and to remove ocular and heart-beat-related components from data.
- Freesurfer was used for segmenting, surface reconstruction, and neuroanatomical labelling based anatomical MRIs. MNE suite was used to create fixed-orientation, cortically constrained surface source models for Morlet-wavelet filtered M/EEG data.
- Source time series were collapsed into fidelity optimized cortical parcels (Korhonen 2014).
- Phase synchronization across all cortical patches was quantified with iPLV and PLV (PLV data not shown here).
- Graph theory (Bullmore 2009) was used to estimate the network structures.
- To correct for multiple tests, we discarded as many significant elements as indicated by the p-value used for the test (0.05 in all tests; discovery discard rate (DDR) correction).
- Parcels were split into r-fcMRI defined functional subsystems for visualization of strongest connections (Yeo 2011). Synchronization was also estimated within and between each subsystem using and its statistical significance was estimated with permutation test (less than 5 % chance).
- Statistical difference between the observed (Hits) and unobserved (Missed) feature changes was tested with t-test ($p < 0.05$, DDR corrected).
- To estimate the correlation of attentional load with the strength of synchronization Pearson's r randomization test was used ($p < 0.05$, DDR corrected).
- To estimate differences between high and low performance, subjects were split into two groups according to hit rate at 4 targets load. Two-way ANOVA ($p < 0.05$, DDR corrected) where the dimensions were Target load x Capacity group was used to estimate interaction between Load and Capacity.

Task and Performance

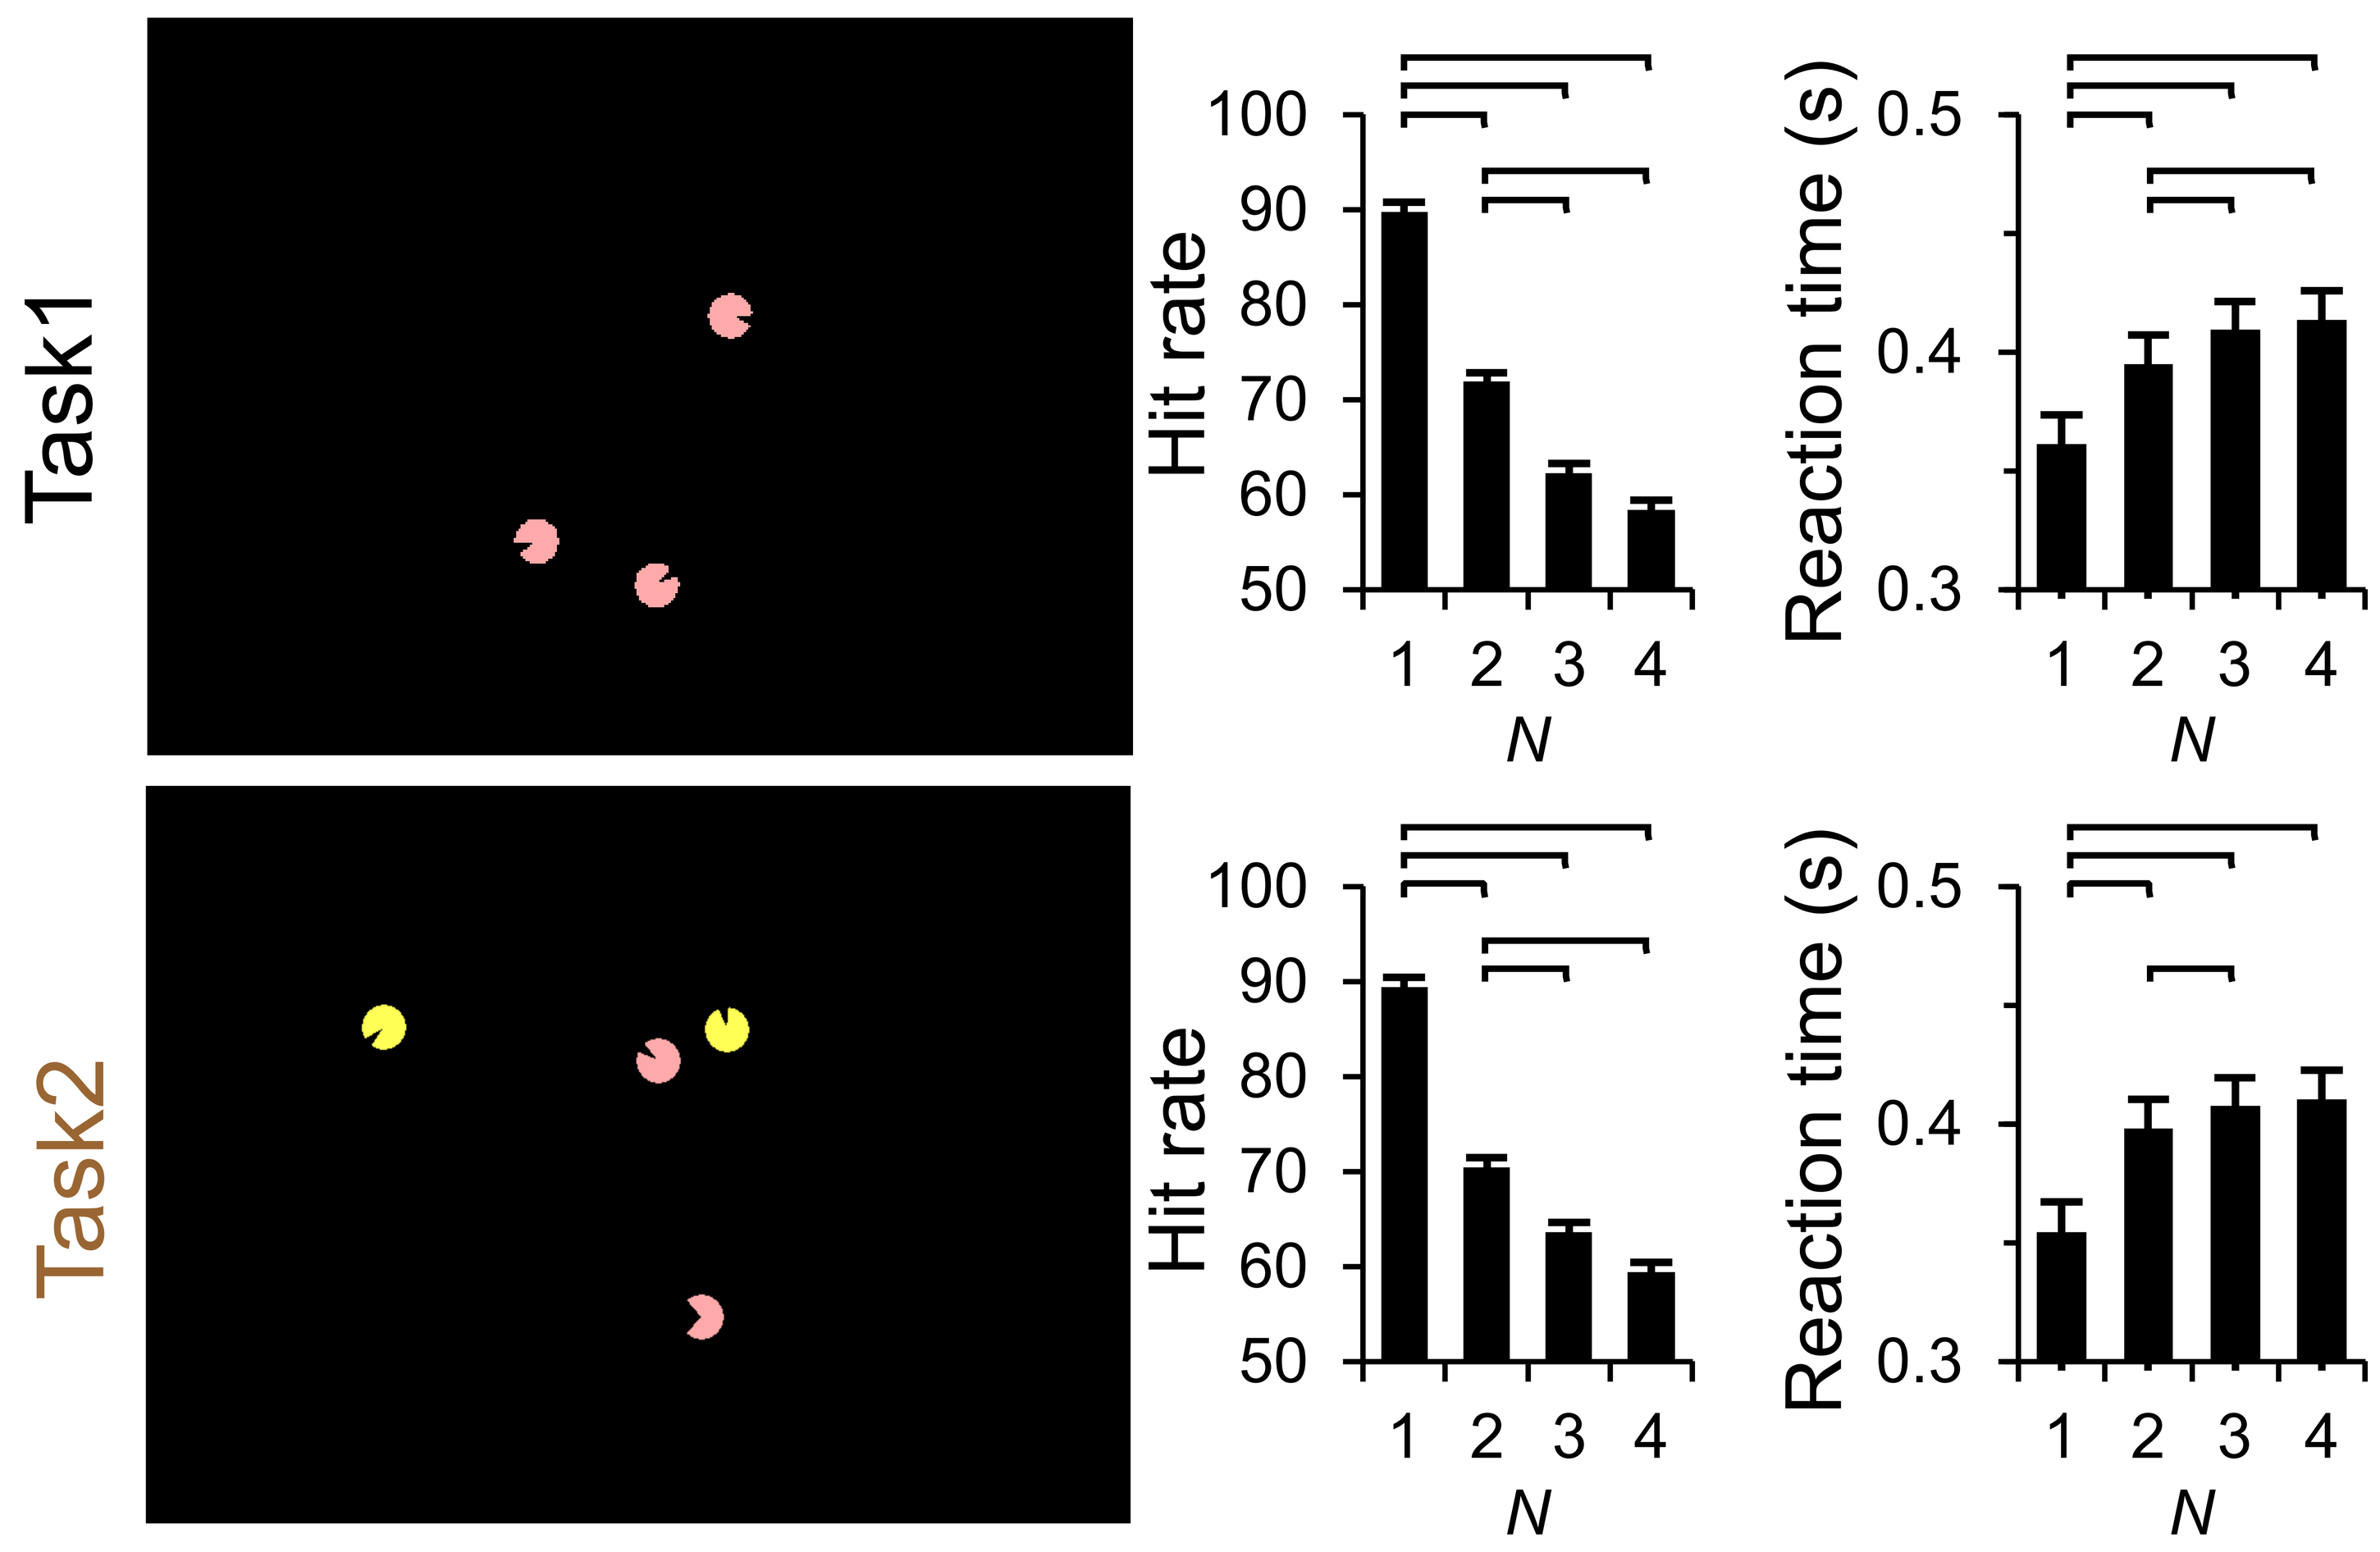
Task1
Task2
Hit rate
Reaction time (s)
N

- Subjects attended 1–4 moving targets and answered when they saw shape changes in the targets.
- No significant difference in hit rates or reaction times was observed between Task1 and Task2 (t-test, $p > 0.1$, uncorrected).
- Bars indicate $p < 0.05$ difference (t-test, Bonferroni corrected).

Behavioral correlates

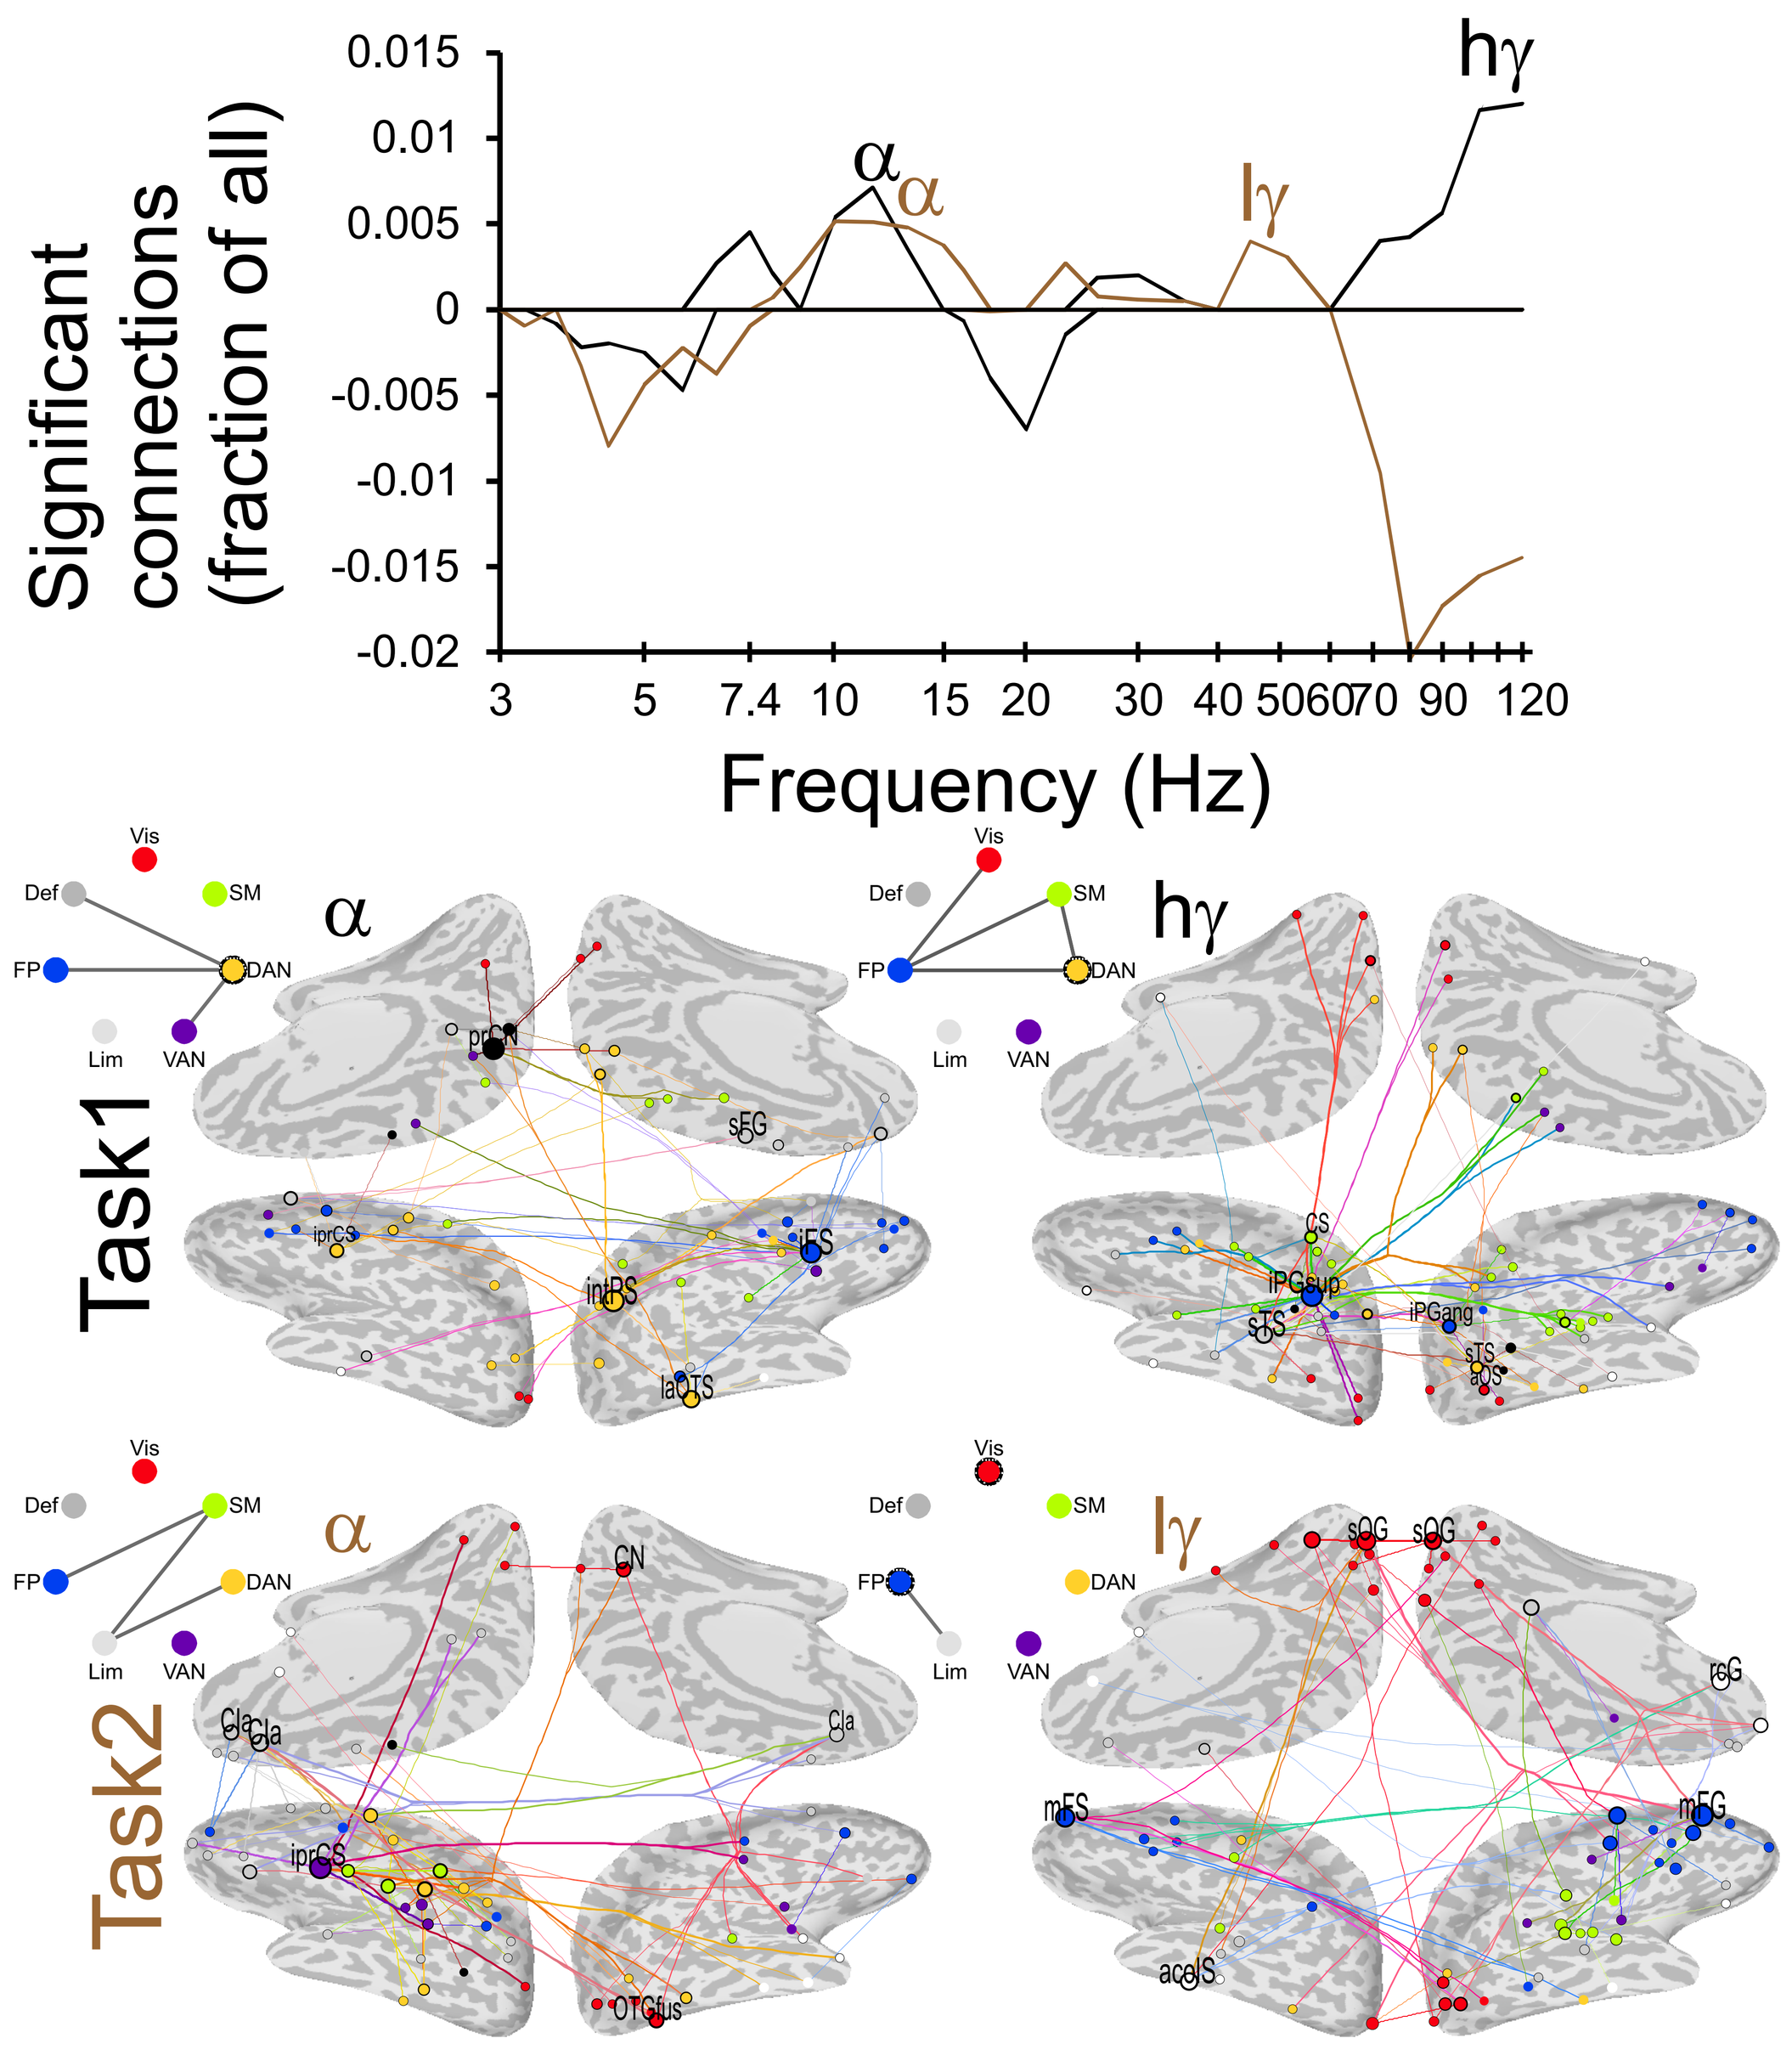
Significant connections (fraction of all)
Frequency (Hz)
Task1
Task2
Vis
SM
Def
FP
DAN
Lim
VAN

- In both tasks synchronization in the high-alpha (10–15 Hz) band was stronger for the detected than for the missed target events and observed in fronto-parietal (FP) and dorsal attention networks (DAN).

High and low capacity subjects have distinct coupling patterns

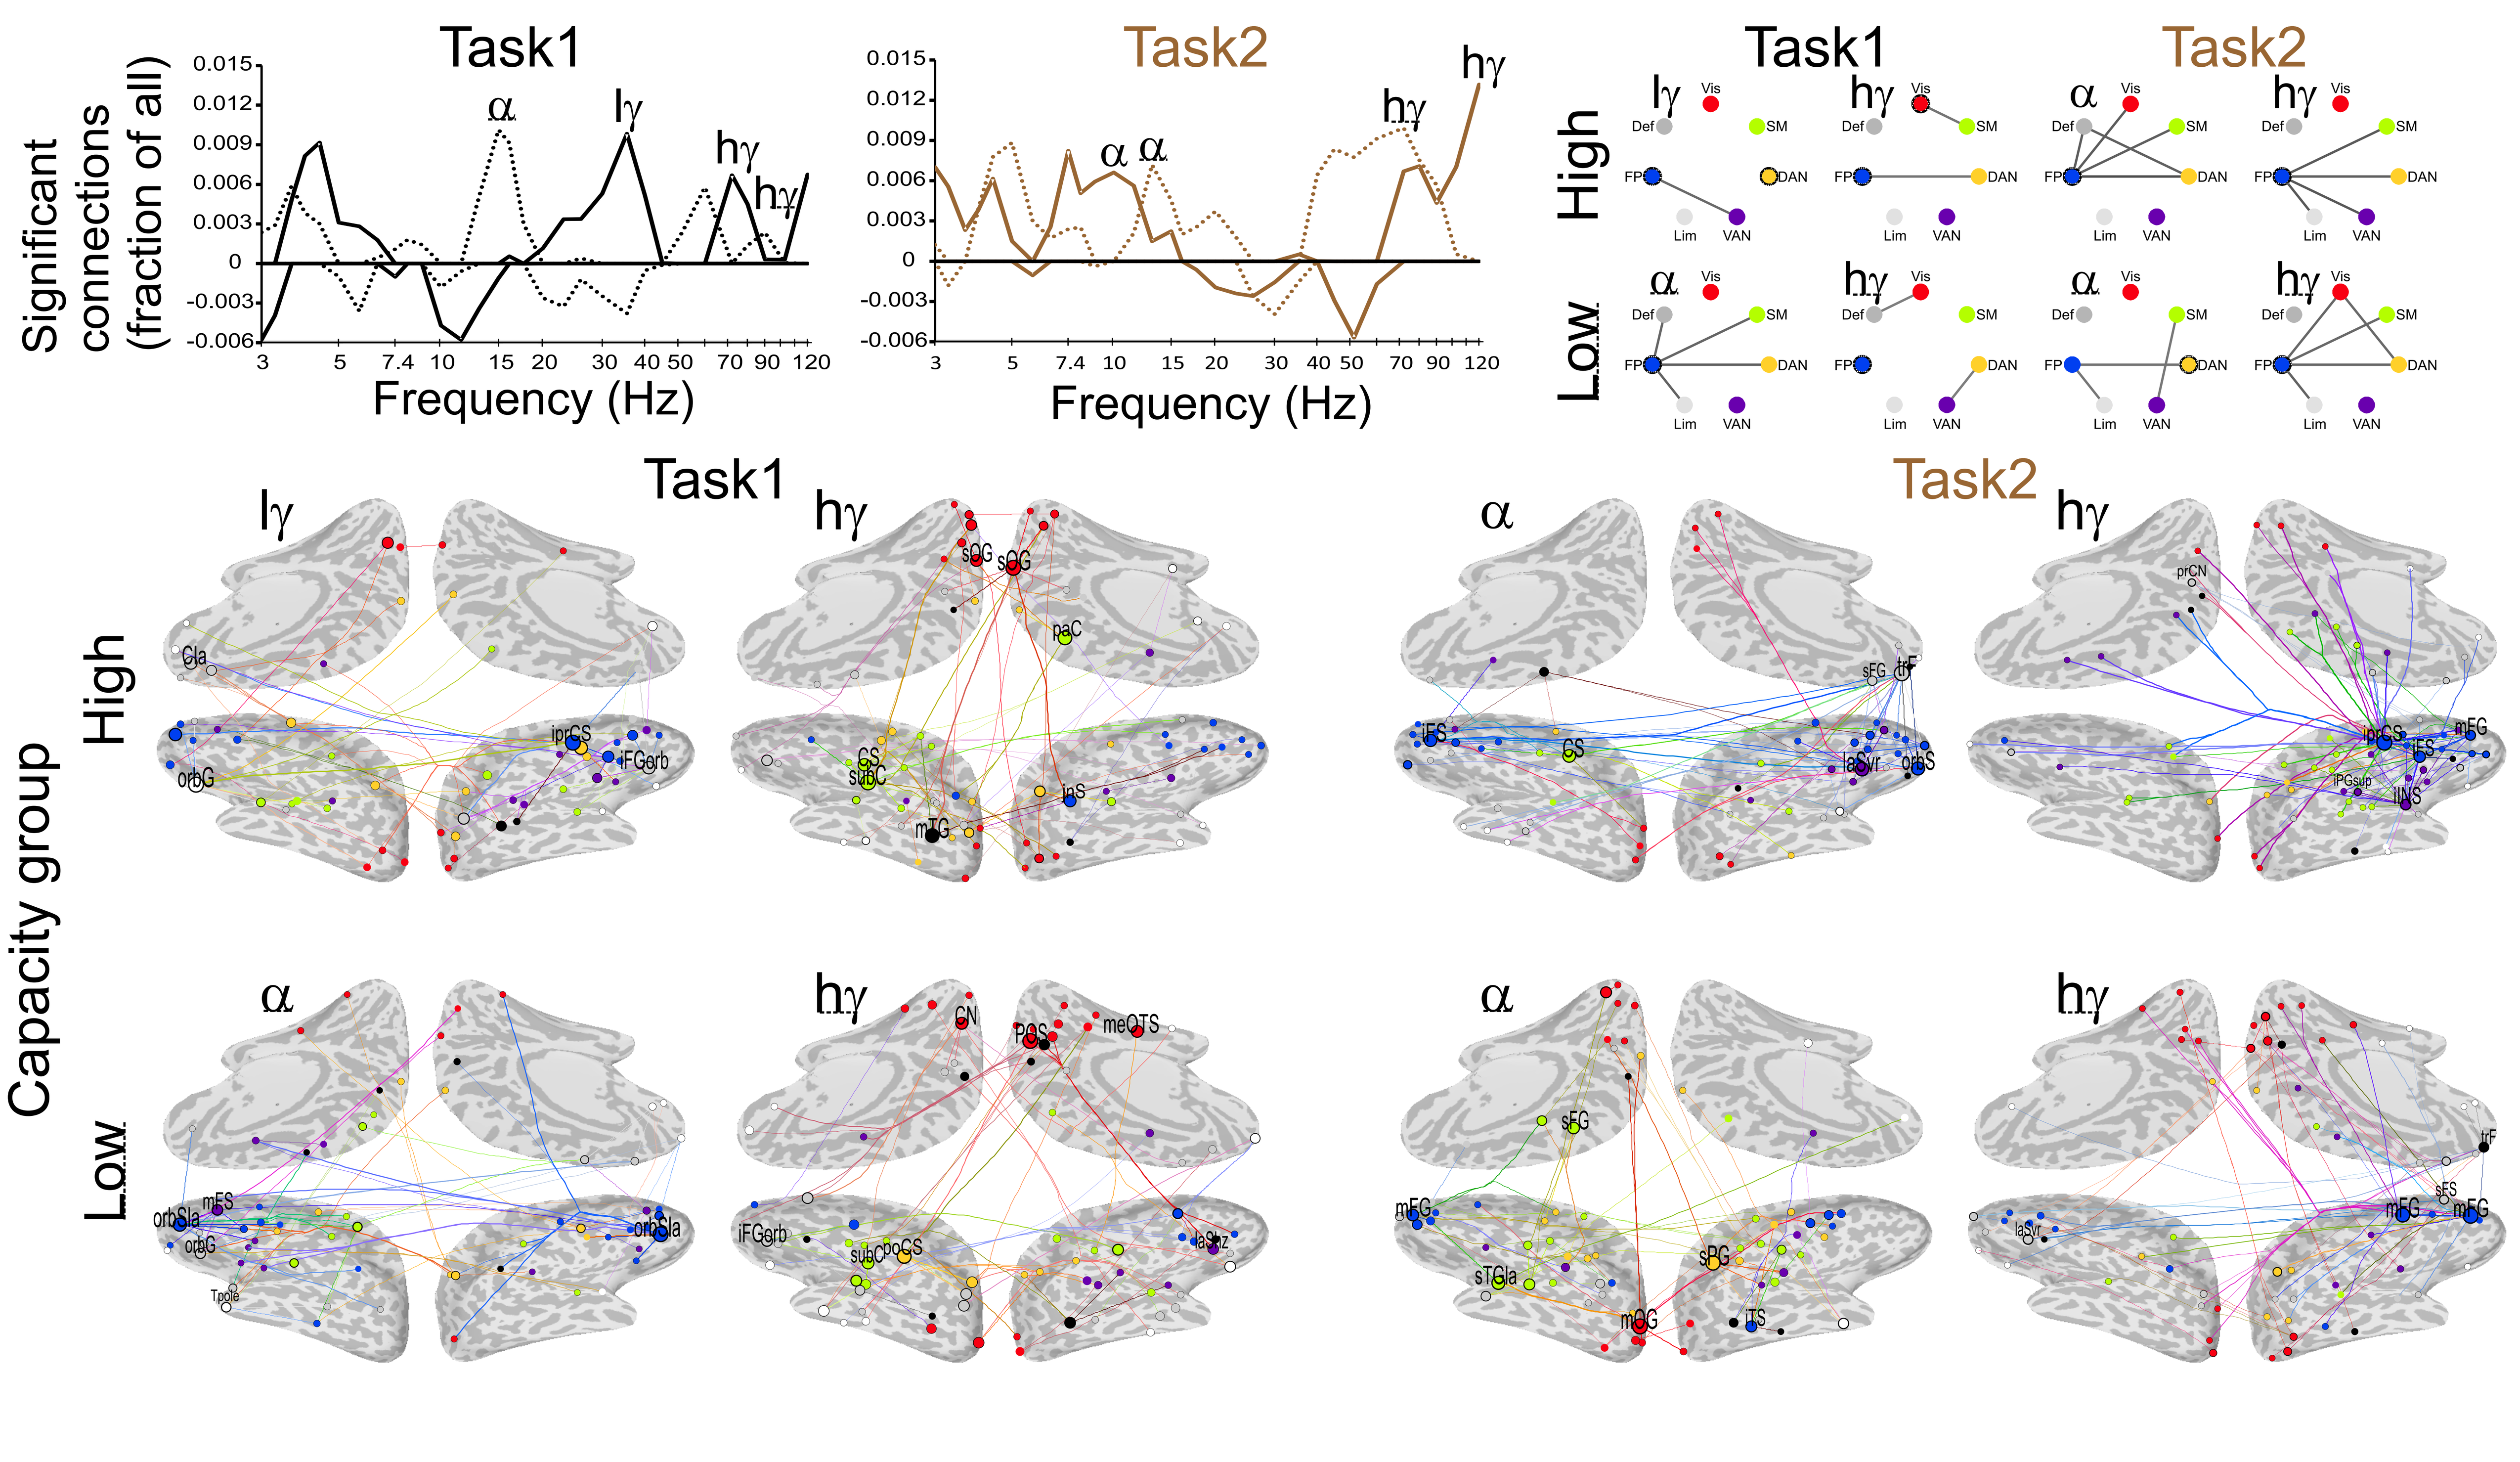
Task1
Task2
Significant connections (fraction of all)
Frequency (Hz)
High
Low
Capacity group

- Synchronization was positively correlated with the number of attended objects in alpha-, beta- (14–30 Hz), and gamma (40–120 Hz) frequency bands.
- Subjects with low and high attentional capacities had distinct spectral and anatomical patterns of load-dependent synchronization.
- There was a significant main load effect and an interaction between load and capacity group in almost all frequencies, but no main effect for the capacity (two-way ANOVA).

Summary

- Large-scale synchronization in FP and DAN networks in high-alpha band predicts correct performance in multi-object attention.
- The capacity of attention is predicted by distinct frequency patterns of synchronization concurrently in alpha, beta, and gamma bands.
- Subjects with high and low attention capacity have distinct connectivity patterns.
- Large-scale synchrony may integrate neuronal processing in FP and DAN and thereby mechanistically support attention to multiple concurrent visual objects.

References

- Bullmore E, Sporns O (2009) Nat Rev Neurosci 10:186-198.
- Culham JC, Brandt SA, Cavanagh P, Kanwisher NG, Dale AM, Tootell RB (1998) J Neurophysiol 80:2657–2670.
- Fries P (2005) Trends Cogn Sci 9:474-480.
- Jovicich J, Peters RJ, Koch C, Braun J, Chang L, Ernst T (2001) J Cogn Neurosci 13:1048 –1058.
- Korhonen O, Palva S, Palva JM (2014) J Neurosci Methods 226C:147-160.
- Oksama L, Hyönä J (2004) Vis Cogn 11:631– 671.
- Rouhinen S, Panula J, Palva JM, Palva S (2013) J Neurosci 33:19023-19033.
- Singer W (1999) Neuron 24:49–65, 111–125.
- Vogel EK, Machizawa MG (2004) Nature 428:748-751.
- Yeo BT, Krienen FM, Sepulcre J, Sabuncu MR, Lashkari D, Hollinshead M, Roffman JL, Smoller JW, Zollei L, Polimeni JR, Fischl B, Liu H, Buckner RL (2011) J Neurophysiol 106:1125-1165.